A new extended-release Topical Formulation of Brimonidine Tartrate – First Clinical Evaluation and Perspectives

J Czernielewski*, G Winckle, * P Andres*

* Tarian Pharma, Grasse, France, www.tarianpharma.com

Introduction and Objectives

TAR-0520 gel is a patent-protected topical formulation of brimonidine tartrate, designed for improved efficacy and 24-hour extended-release of this specific alpha-2 agonist. Applied to the skin, TAR-0520 gel induces local and reversible vasoconstriction of the superficial cutaneous vessels. The phase 1 program evaluated the local tolerance, the PK profile as well as the primary (skin blanching) and secondary (anti-inflammatory) pharmacodynamic activities when TAR-0520 gel was applied to the skin of healthy volunteers (HV).

Materials & Methods

Local tolerance was assessed in a classical 21-day irritation assay with 4 increasing concentrations of TAR-0520 gel (ranging from 0.5% to 1.5%) applied daily to the thorax skin of 20 HV. Application of placebo and of Mirvaso gel served as controls.

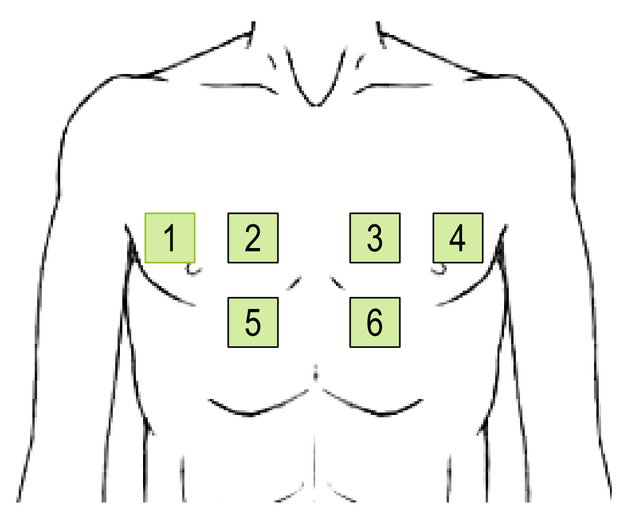
1
2
3
4
5
6

Fig 1: Product application sites

For pharmacokinetic evaluation, 2.0 g of TAR-0520 gel 1% were applied daily to the chest, face and back (1000cm²), of 8 HV for 5 days. Brimonidine level determinations were performed in blood samples collected on day 1 and day 5 using a validated and sensitive analytical method (LOQ 12pg/ml).

The primary pharmacodynamic effect was assessed over 24 h by measuring skin blanching resulting from the superficial vasoconstriction induced by TAR-0520 gel 1.5% in 12 healthy volunteers. A Minolta colorimeter was used to monitor the decrease in the a* value corresponding to the skin blanching intensity.

The secondary pharmacodynamic effect was evaluated in a UV-induced erythema model. After evaluation of the individual minimal erythemal dose (MED), 6 skin areas were exposed to increasing UV doses (1 MED, 2 MED and 3 MED) with 3 areas pretreated (24h and 2h before UV exposure) with TAR-0520 gel and 3 areas pretreated with the same formulation without active. Twenty-four hours after UV exposure, the erythemal response was measured on the 6 areas by clinical observation and by chromametry

Results

No adverse reactions and no SAE were reported during these studies. TAR-0520 gel was shown to be well tolerated with no sign or symptom of irritation when applied at concentrations up to 1.5% (see Table 1 below)

Products	TAR-0520 gel 0,5%	TAR-0520 gel 0.75%	TAR-0520 gel 1%	TAR-0520 gel 1.5%	Reference gel 0,5%	TAR-0520 gel vehicle
Cumulative Irritation score	0.05	0	0.1	0	0.1	0.05
Total subjects n/%	20 (100%)	20 (100%)	20 ((100%)	20 (100%)	20 (100%)	20 (100%)
No irritation % score 0	19 (95%)	20 (100%)	19 (95%)	20 (100%)	19 (95%)	19 (95%)
Minimal erythema score 1	1 (5%)	0	1 (5%)	0	1 (5%)	1 (5%)
Mild irritation or worse score 2+	0	0	0	0	0	0

The PK assessment conducted under maximized conditions confirmed a slow and extended release of the active with the systemic exposures similar to those reported with marketed products that contain lower brimonidine concentrations. See Table 2 below for AUC 0-24h comparisons.

Product	Alphagan 0.2%	Mirvaso 0.5%	TAR-0520 1%
Regimen	TID	QD	QD
AUC 0-24h (pg*h/ml)	568 ± 277	417 ± 263	554 ± 448
Range	124-1490	10-1077	185-1806
Accumulation ratio	1.9	1.2	1.7

The primary pharmacodynamic effect confirmed a dose dependent and long-lasting TAR-0520 gel activity, with the highest concentrations showing vasoconstriction (skin blanching) over a period of 24 hours (Figure 2).

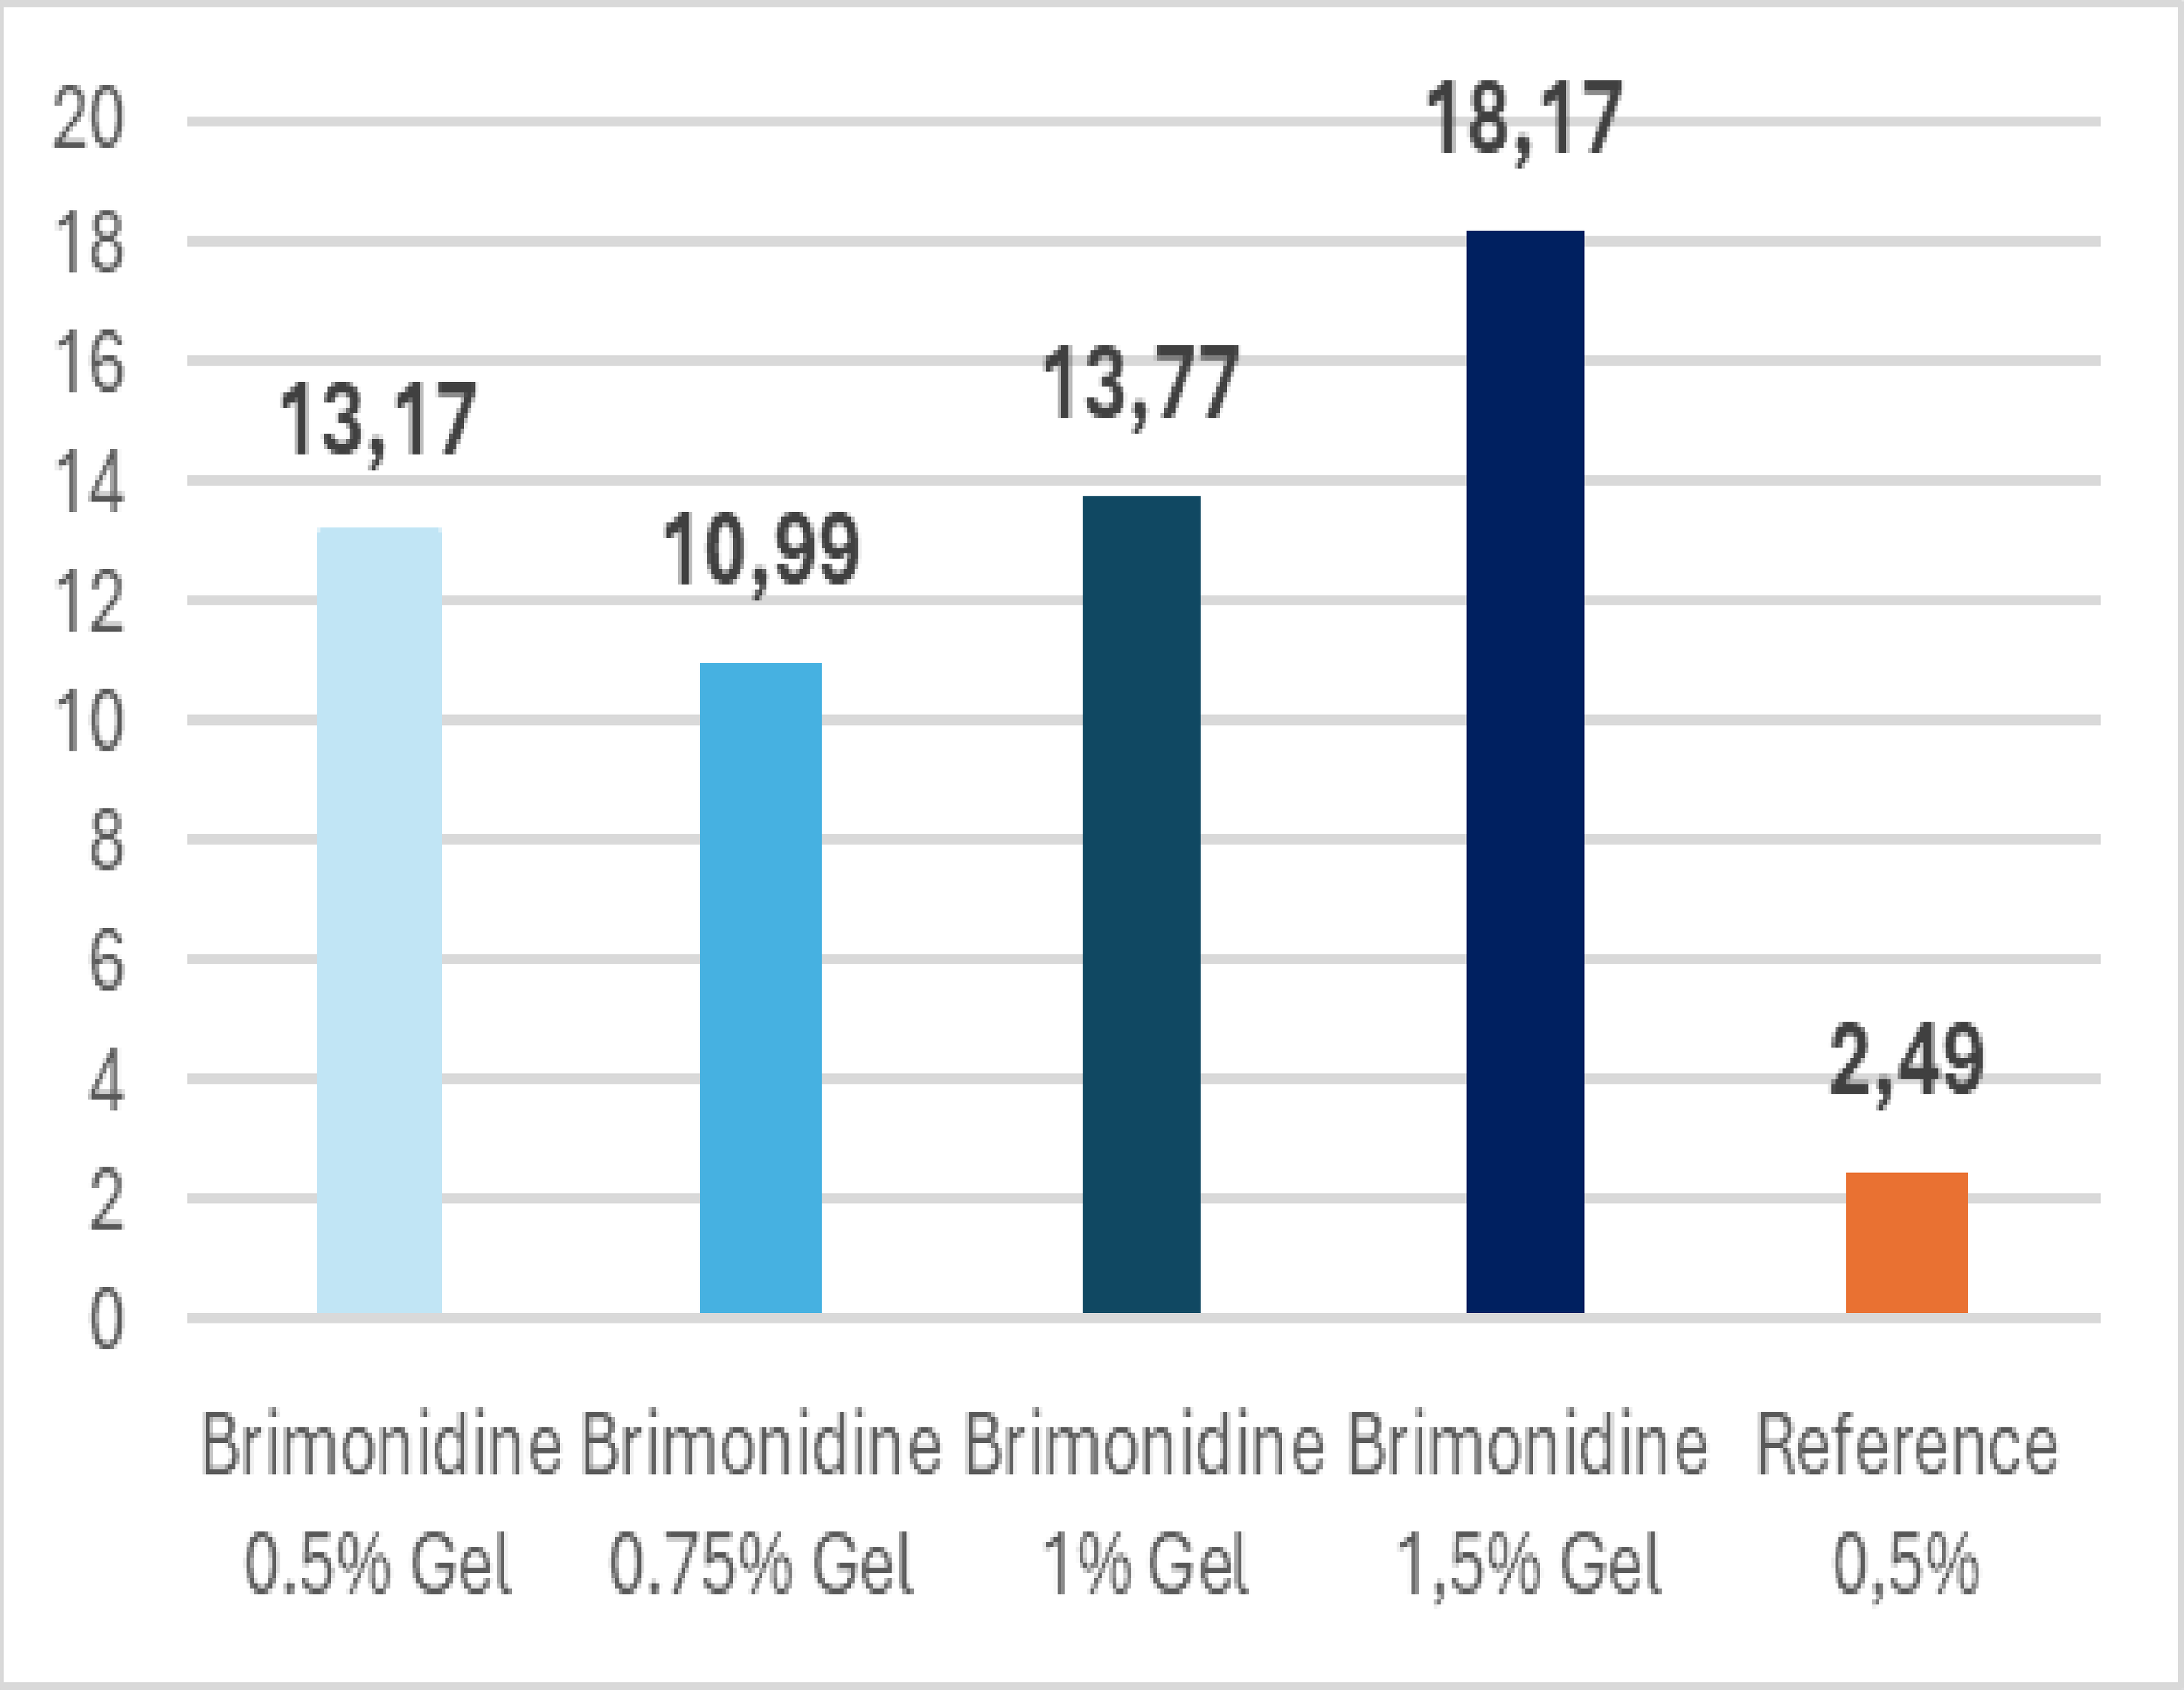
20
18
16
14
12
10
8
6
4
2
0
13,17
10,99
13,77
18,17
2,49
Brimonidine 0.5% Gel
Brimonidine 0.75% Gel
Brimonidine 1% Gel
Brimonidine 1,5% Gel
Reference 0,5%

Figure 2: AUC of delta a* (versus untreated) from 0 to 24 hours

The secondary pharmacodynamic effect was demonstrated with a significant erythema reduction in the area treated with TAR-0520 gel: 100% erythema reduction observed at MED x 1 and at MED x 2 and 70% erythema reduction at MED x 3, (Figure 3).

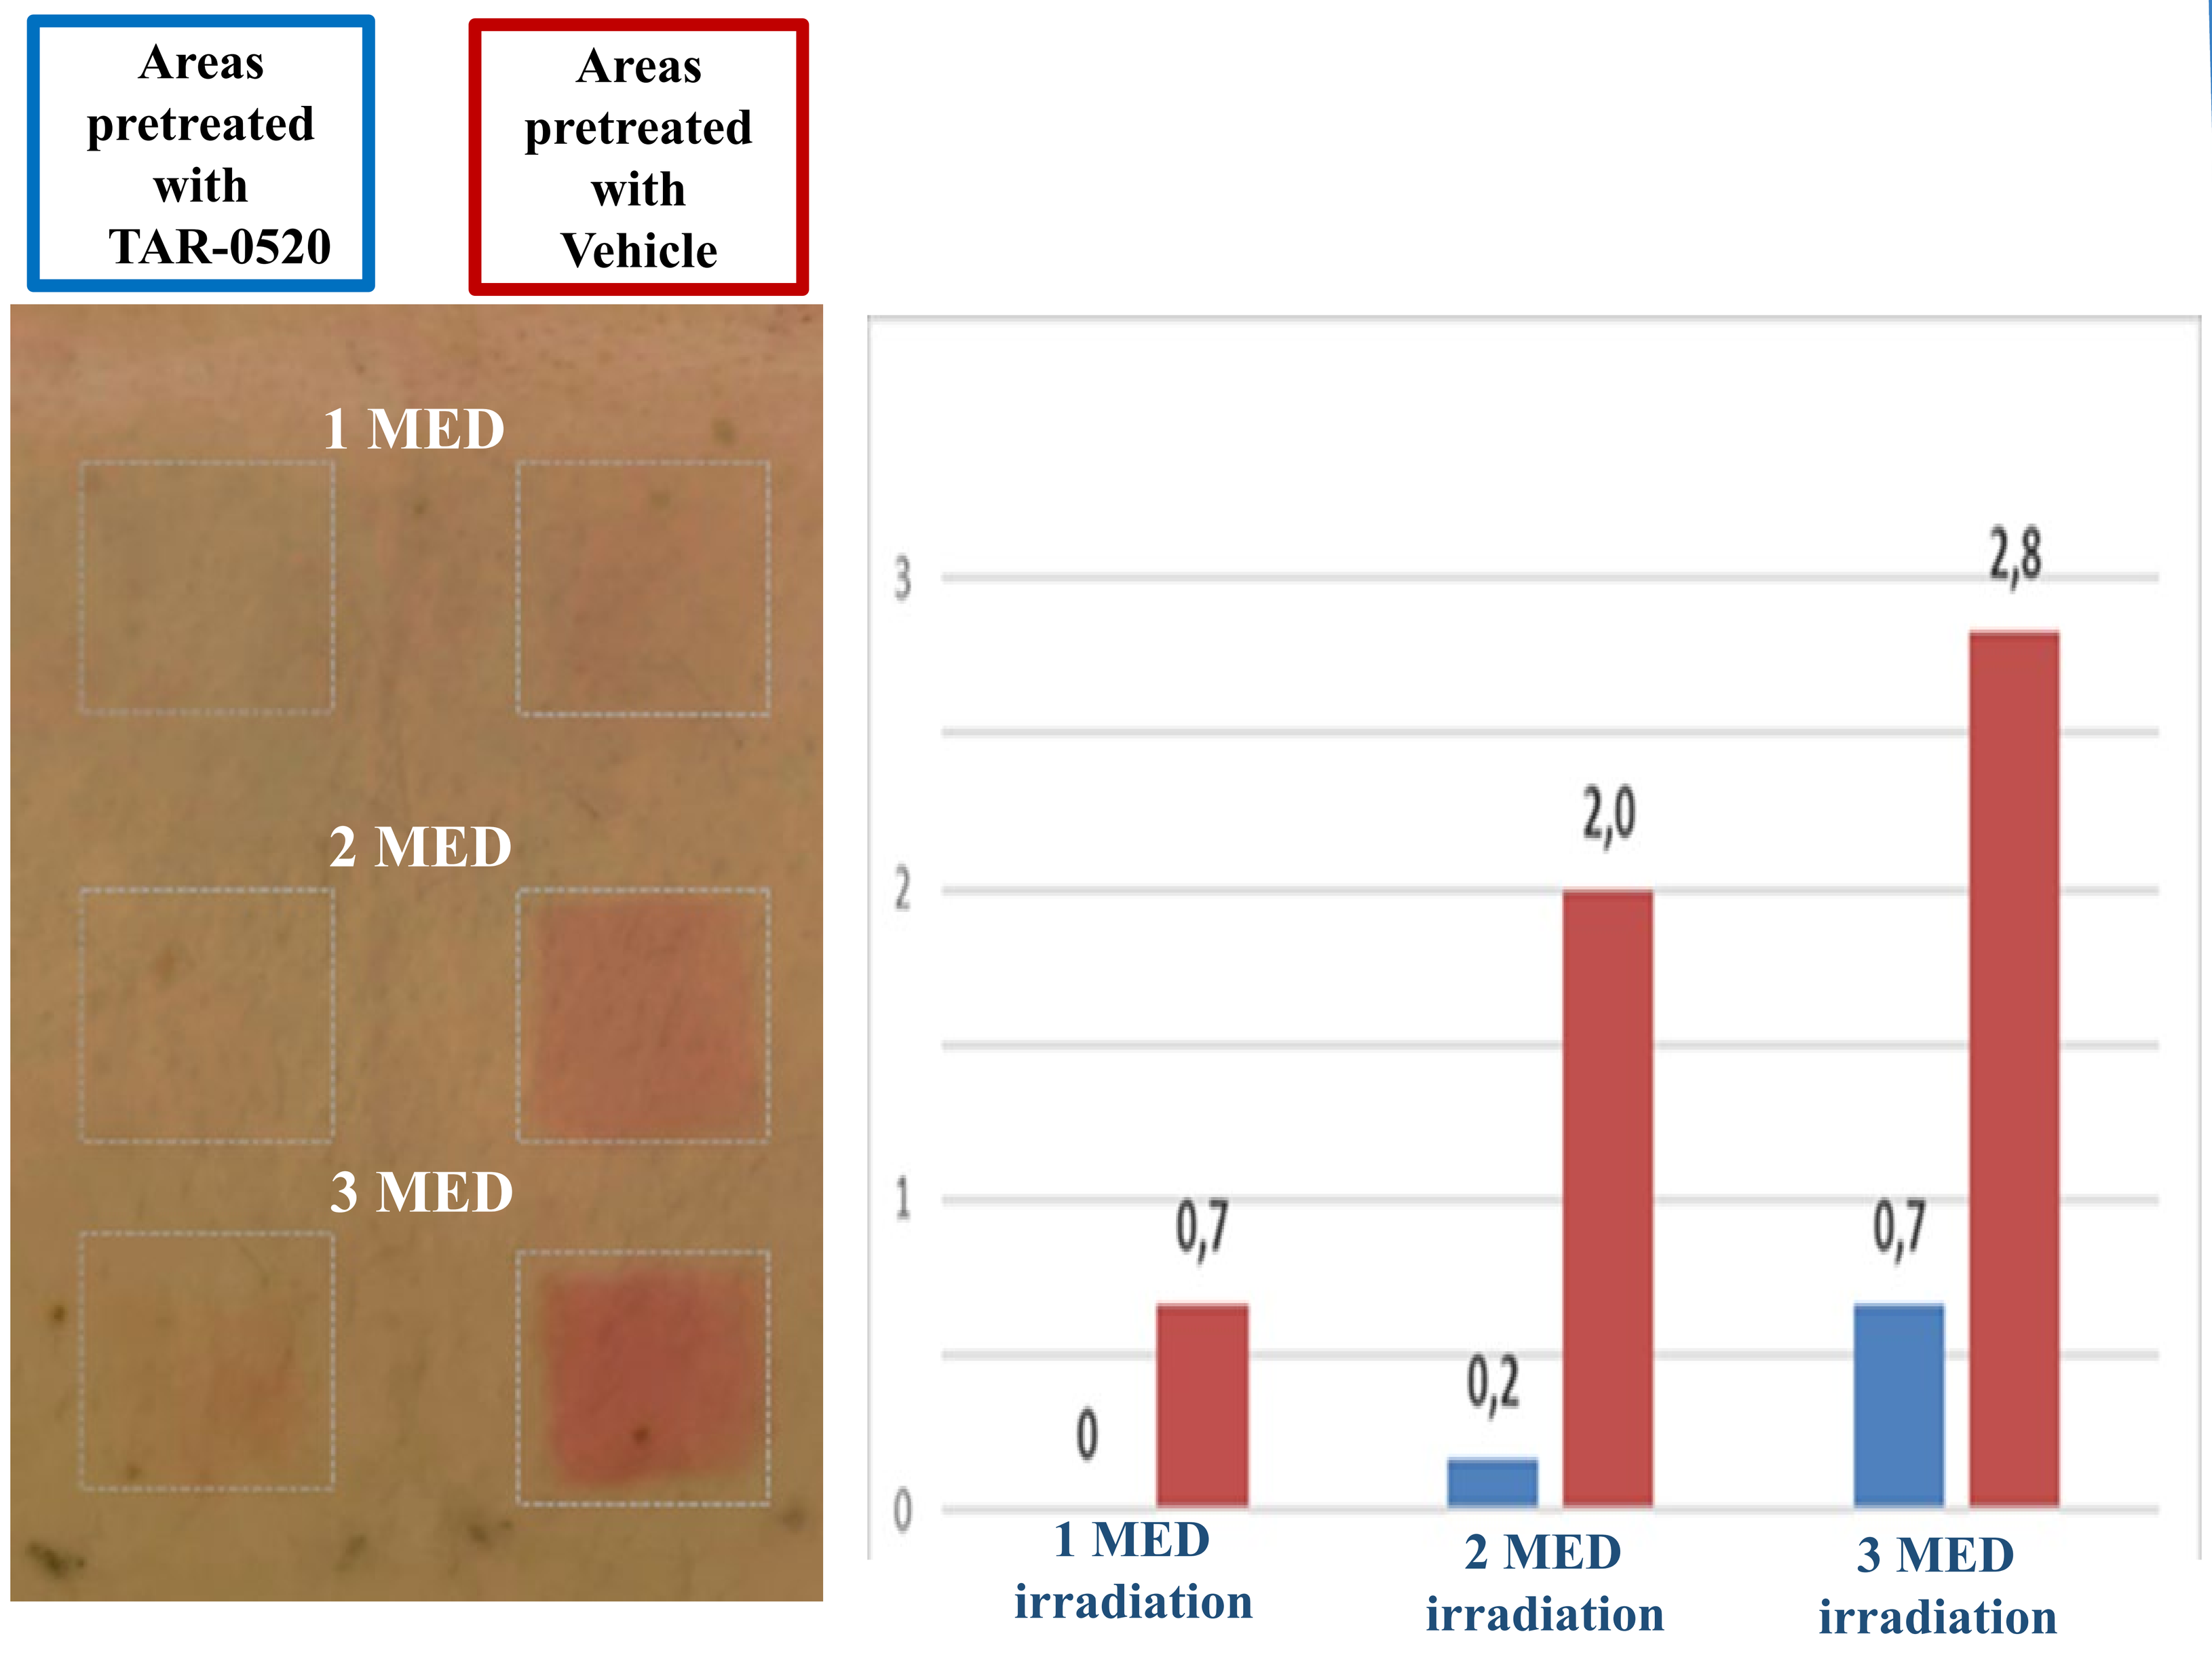
Areas pretreated with TAR-0520
Areas pretreated with Vehicle
1 MED
2 MED
3 MED
3
2
1
0
0
0,7
0,2
2,0
0,7
2,8
1 MED irradiation
2 MED irradiation
3 MED irradiation

Figure 3. Mean clinical erythema score at Day 3 (0 to 4 scale). Skin zones were pretreated with TAR-0520 gel (blue) or placebo (red) , 24h & 2h before UV exposure.

Conclusions

In a clinical evaluation involving healthy volunteers, TAR-0520 gel demonstrated excellent local tolerability and minimal systemic absorption, underscoring a favorable safety profile. The sustained vasoconstrictive response, in combination with potent anti-inflammatory activity, positions TAR-0520 gel as a promising topical intervention for the prevention of chemotherapy-induced skin toxicities and the management of chronic inflammatory skin diseases.

References

1. Bertino B, Blanchet-Réthoré S, Thibaud de MénonvilleS et all. Brimonidine displays anti-inflammatory properties in the skin through the modulation of the vascular barrier function. Exp Dermatol. 2018;27(12):1378-1387

2. Piwnica D,Rosignol C,Thibaut de Menonville S et all, Vasoconstriction and anti-inflammatory properties of the selective a-adrenergic receptor agonist brimonidine. J Dermatol Science 2014; 75:49-54